IRL201104, A Novel Immunomodulatory Peptide, Shows Efficacy on Inflammatory Endpoints and Airway Hyperresponsiveness in a House Dust Mite Driven Model of Allergic Inflammation With or Without Poly I:C Exacerbation

J De Alba, L Ravanetti, P Eggleton, V Corrigall and R Foulkes

Revolo Biotherapeutics Ltd Gaithersburg, USA/London, UK

Introduction

- IRL201104 (1104) is a novel clinical stage immunomodulatory peptide derived from an mTB chaperonin which, despite a very short half-life (~10-15min across species), shows a long-lasting effect in models of T2 allergic inflammation (Riffo-Vasquez et al, 2019; Page et al, 2019, De Alba et al, 2022).
- The aim of the study was to explore the effect of 1104 in a previously described allergic inflammation model (De Alba et al, 2015) driven by house dust mite (HDM) in Freund's complete adjuvant (FCA) characterized by a mixed T cell phenotype and in the presence or absence of the viral mimetic Polyinosinic-polycytidylic acid (Poly I:C). Furthermore, we have also studied for the first time the effect of 1104 on airway hyperresponsiveness (AHR) measured by Resistance/Compliance.

Methods

Model

- All mice were sensitised with HDM (100 µg, s.c., GREER) in FCA (Sigma) at day 1 and then intranasally challenged at day 14 with either HDM (25 µg) or saline. A second group of animals also received high molecular mass Poly I:C (30 µg/animal) or saline administered intranasally 24h before HDM/saline challenge. All endpoints were taken 48h after HDM challenge, other than AHR which was measured 24h after challenge.
- 1104 (iv; 80µg/kg) or vehicle was administered once 15min before allergen challenge. Dexamethasone (po;1 mg/kg) was administered 1h before allergen challenge.

BALF collection, differential cell counts and cytokine/chemokine measurements

- 48h after HDM challenge, animals were overdosed with pentobarbitone and bronchoalveolar lavage was carried out using phosphate buffered saline. The isolated bronchoalveolar lavage fluid (BALF) was then centrifuged at 1500 rpm for 10min at 4ºC and the supernatant was aliquoted and stored at -80ºC for cytokine analysis. Cell pellets were then re-suspended in 0.2% w/v NaCl to induce haemolysis of any erythrocytes. After isotonization with the same volume of 1.6% w/v NaCl, the BALF cells were analysed for total and differential cell numbers using a XT-2000iV analyser (Sysmex).
- A 14-Plex cytokine/chemokine panel (IFN-ɣ,IL-4, IL-5, IL-6, KC, IL-10, IL-12(p40), IL-13, IL-17, Eotaxin, G-CSF, GM-CSF, RANTES and MCP-1) was run in BALF supernatant using a magnetic multiplex assay as per the manufacturer's instructions (Biotechne Ltd). Levels were measured using a Magpix system (Luminex Corp).

HDM specific IgE ELISA assay

- 48h after HDM challenge a terminal blood sample was collected via cardiac puncture and serum separated. HDM specific IgE concentration in serum was determined using an ELISA kit (Condrex Inc.) as per the manufacturer's instructions. Optical density was measured at 450 nM using a microplate reader (SpectraMax 340PC). Concentrations of IgE were determined using SoftMax Pro v. 6.4 (Molecular Devices).

Assessment of airway hyper-responsiveness (AHR)

- 24h after HDM challenge, mice were anaesthetised and tracheotomized. After surgery, they were placed in a whole-body plethysmograph and the tracheotomy tube was connected to a mechanical ventilator (rate 150 breaths/minute; tidal volume 0.15 – 0.2 mL). Flow signal was recorded using the plethysmograph and pressure signal was recorded from a sidearm of the tracheal catheter. Flow and pressure signals were processed together to determine lung resistance (R_L) and dynamic compliance (Cdyn) using a software analyser provided in the FinePointe resistance–compliance software (DSI Inc.).
- After 10min stabilization, initial baseline readings were taken. Bronchoconstriction was then evoked with 10µL aerosolised methacholine (MCh 3, 10 and 30 µg/ml for 20 s). Changes to Cdyn and R_L were calculated from the difference between the baseline level (20 breath averaged before challenge) and maximum effect below or above baseline level within 5 min of challenge.

Statistical analysis

- Data are shown as mean ± S.E.M. (standard error of the mean). Inter-group deviations were statistically analysed by a one-way analysis of variance (ANOVA) followed by a Dunnett's test or a Student's t-test when comparing HDM/saline vs HDM/Poly I:C groups. P< 0.05 was considered statistically significant.

Results (I)

Effect of treatment on HDM induced cell infiltration in the presence/absence of Poly I:C exacerbation

48h post-challenge, exposure to intranasal HDM, triggered inflammatory infiltration in the lung as measured by BALF differential cell counts. Surprisingly, as this model is usually refractory to steroids, both dexamethasone and 1104 significantly reduced HDM-induced lung infiltration of eosinophils, neutrophils, and lymphocytes but did not affect macrophages (Fig1).

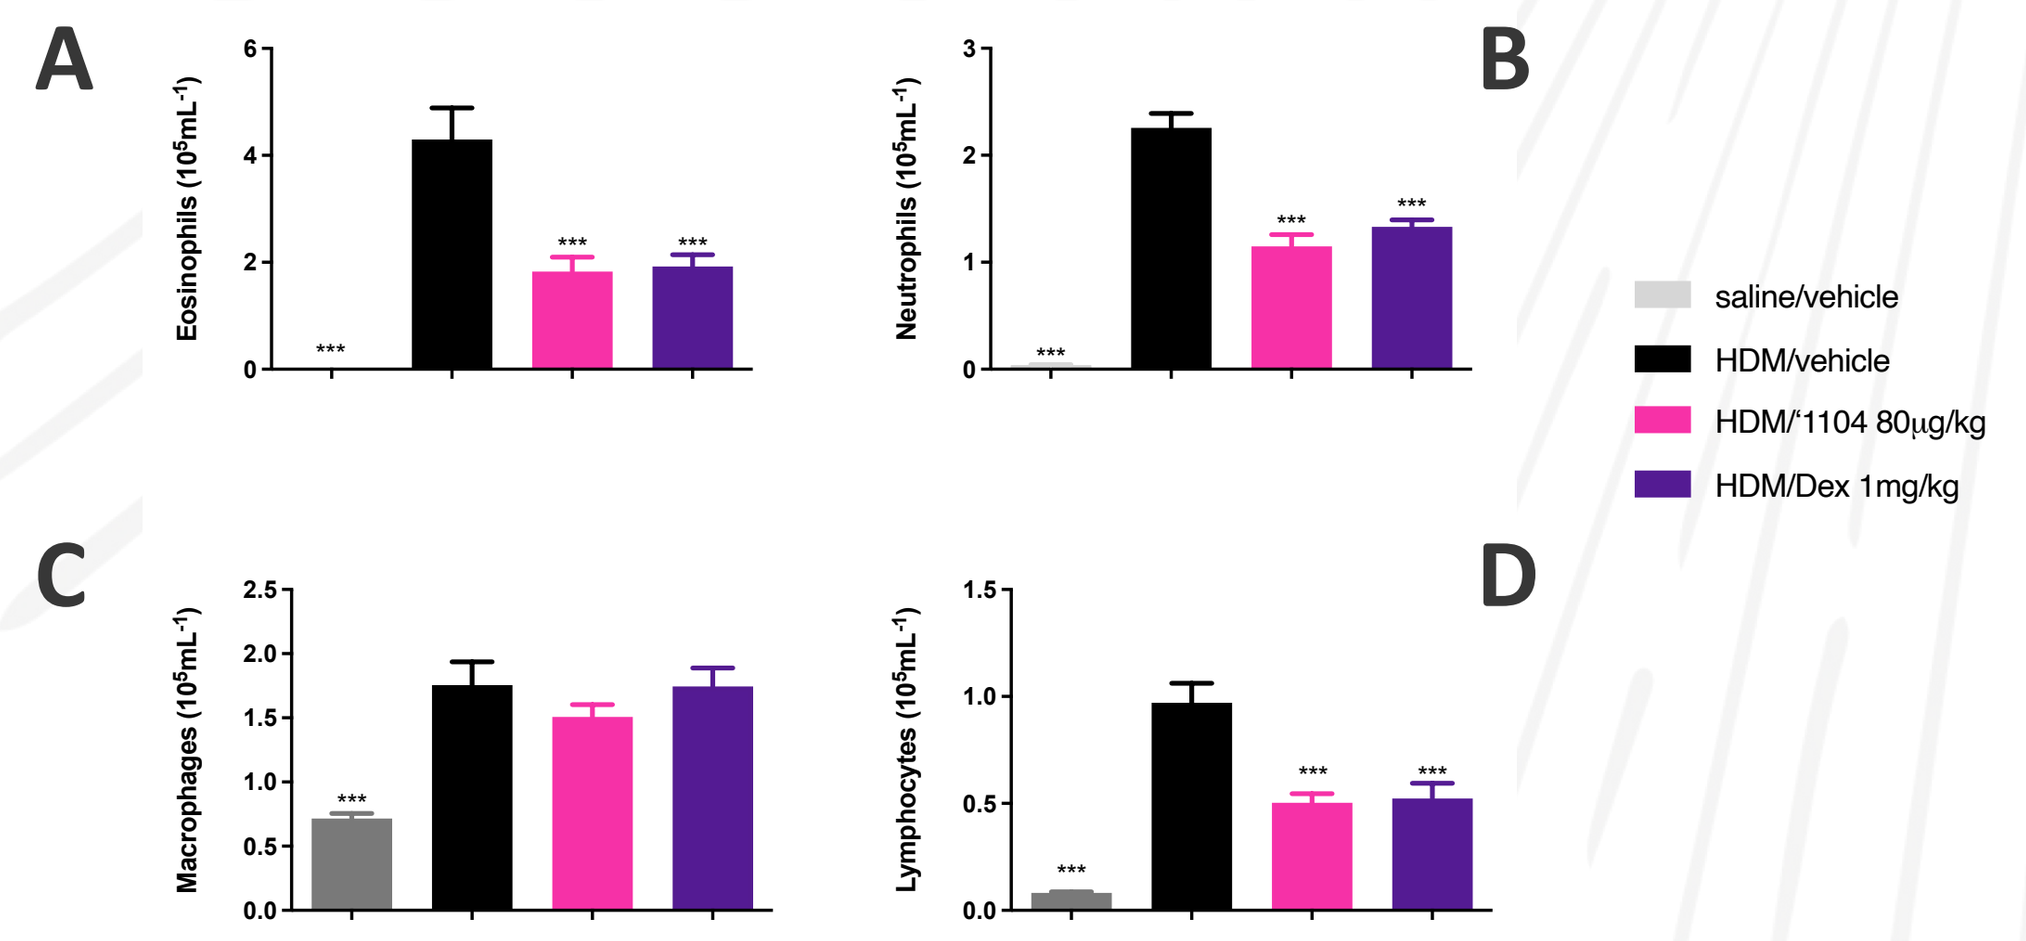
A
B
C
D
Eosinophils (10⁵mL⁻¹)
Neutrophils (10⁵mL⁻¹)
Macrophages (10⁵mL⁻¹)
Lymphocytes (10⁵mL⁻¹)
saline/vehicle
HDM/vehicle
HDM/'1104 80µg/kg
HDM/Dex 1mg/kg

Figure 1 – Effect of treatment on HDM-induced cellular infiltration. *Effect on (A) eosinophils, (B) neutrophils, (C) macrophages and (D) lymphocytes lung infiltrate. Data are expressed as cells per mL of BALF, mean ± SEM. Groups were compared to HDM/vehicle group using a one-way ANOVA, followed by a Dunnett's test,***P<0.001; n=8.*

Exposure to Poly I:C 24 before HDM challenge, elicited exacerbated inflammatory infiltration which was significant in macrophages and lymphocytes (Fig2C,D). Dexamethasone caused a significant inhibition of eosinophils, while 1104 significantly reduced both eosinophilic and neutrophilic infiltration (Fig2A,B).

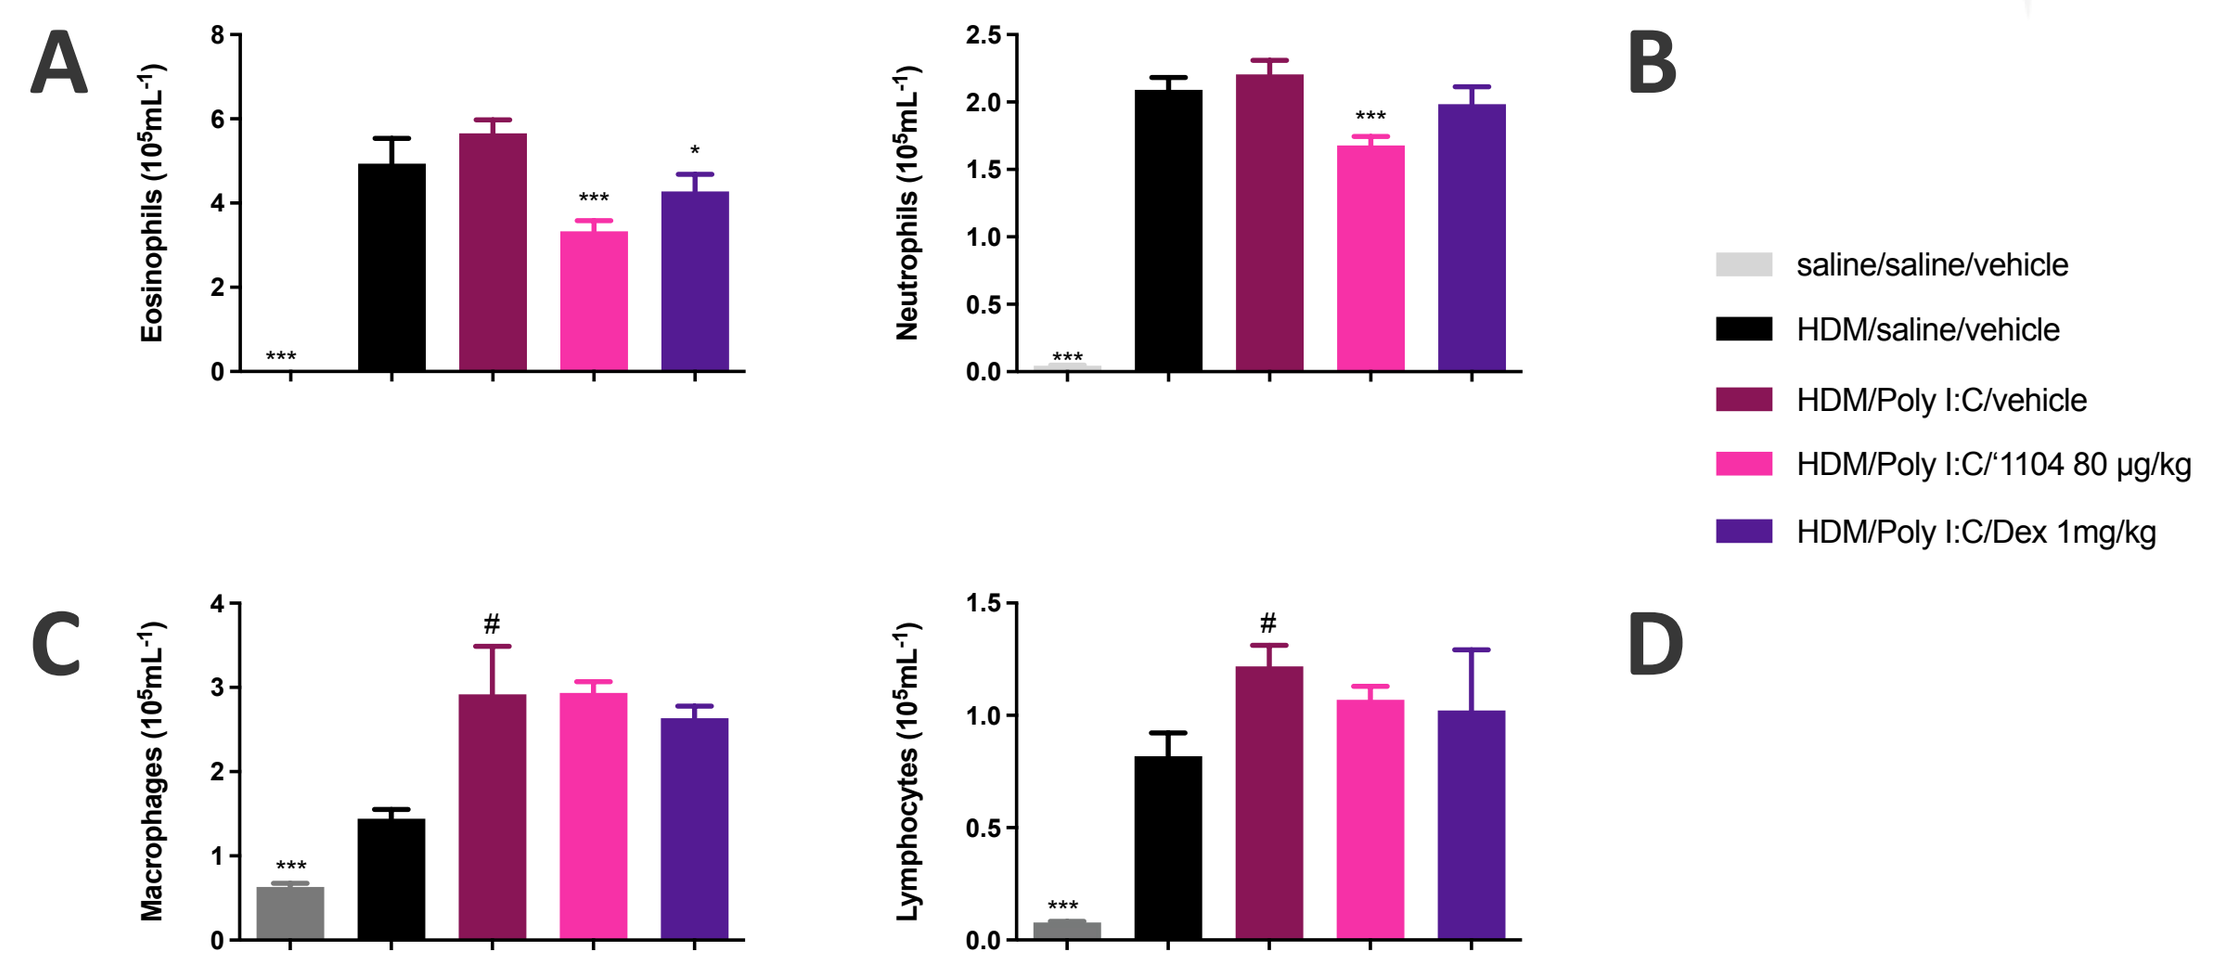
A
B
C
D
Eosinophils (10⁵mL⁻¹)
Neutrophils (10⁵mL⁻¹)
Macrophages (10⁵mL⁻¹)
Lymphocytes (10⁵mL⁻¹)
saline/saline/vehicle
HDM/saline/vehicle
HDM/Poly I:C/vehicle
HDM/Poly I:C/'1104 80 µg/kg
HDM/Poly I:C/Dex 1mg/kg

Figure 2 – Effect of treatment on HDM-induced cellular infiltration exacerbated by Poly I:C exposure. *Effect on (A) eosinophils, (B) neutrophils, (C) macrophages and (D) lymphocytes lung infiltrate. Data are expressed as cells per mL of BALF, mean ± SEM. Groups were compared to HDM/Poly I:C/vehicle group using a one-way ANOVA, followed by a Dunnett's test;*P<0.05,***P<0.001. A Student's t-test was used when comparing HDM/saline/vehicle vs HDM/Poly I:C/vehicle group;#P<0.05; n=8.*

Effect of treatment on HDM specific IgE in serum

HDM challenge elicited an increase in HDM specific IgE in serum with was exacerbated by Poly I:C exposure (Fig3A,B). Dexamethasone treatment significantly reduced HDM specific IgE in serum in the non exacerbated model (Fig3A) while 1104 was effective in both the exacerbated and non exacerbated model (Fig3A,B)

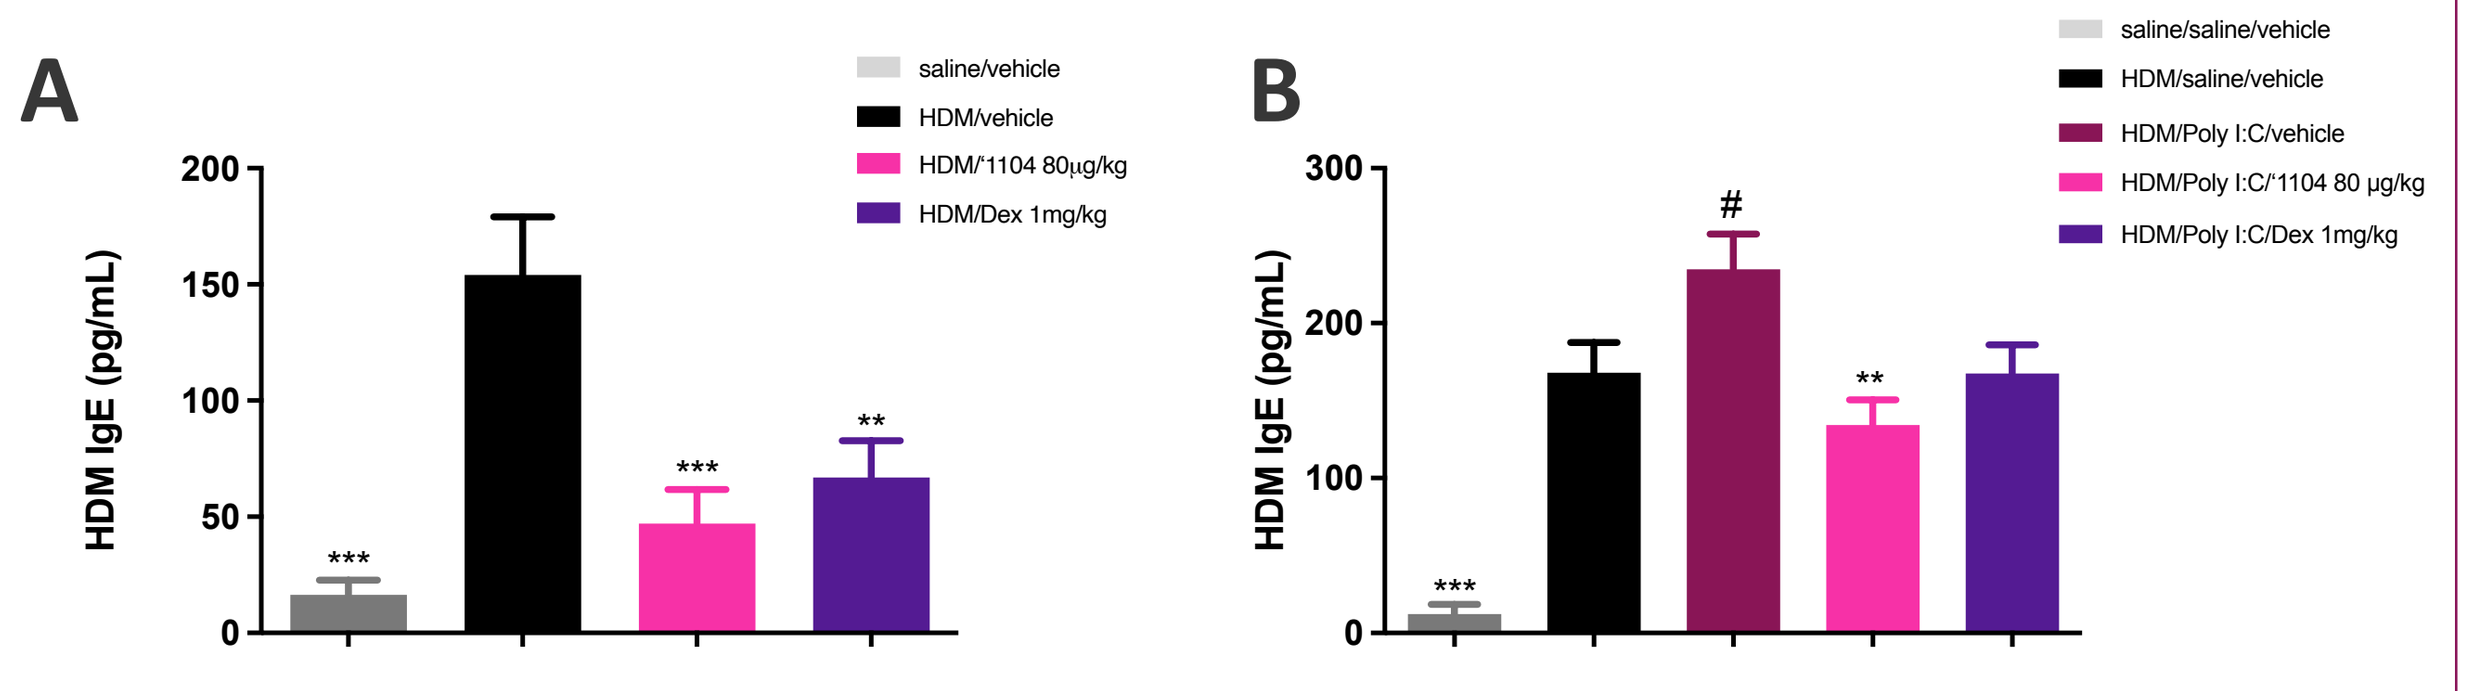
A
B
HDM IgE (pg/mL)
saline/vehicle
HDM/vehicle
HDM/'1104 80µg/kg
HDM/Dex 1mg/kg
saline/saline/vehicle
HDM/saline/vehicle
HDM/Poly I:C/vehicle
HDM/Poly I:C/'1104 80 µg/kg
HDM/Poly I:C/Dex 1mg/kg

Figure 3 – Effect of treatment on serum HDM specific IgE. *Effect of Dexamethasone and 1104 on serum HDM specific IgE in the absence (A) or presence (B) of Poly I:C exacerbation. Data are expressed as pg/mL, mean ± SEM. Groups were compared to either to HDM/vehicle (A) or HDM/Poly I:C/vehicle (B) using a one-way ANOVA, followed by a Dunnett's test, ;**P<0.01,***P<0.001. A Student's t-test was used when comparing HDM/saline/vehicle vs HDM/Poly I:C/vehicle group;#P<0.05; n=8.*

Results (II)

Effect of treatment on cytokine/chemokine release in BALF

48h after allergen challenge, HDM alone or in combination with Poly I:C elicited the release of IFN-ɣ, IL-4, IL-5, IL-6, KC, IL-10, IL-12(p40), IL-13, IL-17, Eotaxin, G-CSF, GM-CSF, RANTES and MCP-1 in BALF supernatant. Both dexamethasone and 1104 significantly suppressed these cytokines/chemokines except for IL-10, IL-17 and RANTES (Table 1). Poly I:C exposure 24h before HDM challenge significantly exacerbated the release of IFN-ɣ, IL-5, IL-6, KC, IL-13, G-CSF, RANTES and MCP-1. As expected, the exacerbated model was largely refractory to steroids, with dexamethasone only significantly impacting IL-5 and IL-6. In contrast 1104 retained a significant effect on IFN-ɣ, IL-4, IL-5, IL-6, KC, IL-13, Eotaxin and MCP-1 (Table 2).

BALF (pg/ml)	Saline/vehicle	HDM/vehicle	HDM/1104 80µg/Kg i.v.	HDM/Dex 1mg/kg p.o.
IFN-ɣ	13±7***	363±35	121±18***	190±28***
IL-4	19±7***	495±39	148±22***	206±23***
IL-5	18±7***	416±30	122±19***	175±20**
IL-6	11±6***	436±36	101±19***	169±21***
IL-10	13±6.5*	86±17	73±18	74±20
KC	20±8***	299±27	111±16***	151±23***
IL-12p40	20±8***	254±21	144±22**	180±23
IL-13	19±7***	303±29	90±11***	131±15***
IL-17	13±6***	89±12	62±11	77±11
Eotaxin	18±7***	267±26	100±19***	123±20***
G-CSF	12±6***	217±21	113±14***	137±20**
GM-CSF	20±8***	180±19	76±14***	106±20**
RANTES	14±7	40±14	27±11	28±11
MCP-1	12±6***	269±29	112±20***	159±22**

Table 1– Effect of treatment on cytokines/chemokines in BALF after HDM challenge. *Effect on IFN-ɣ, IL-4, IL-5, IL-6, KC, IL-10, IL-12(p40), IL-13, IL-17, Eotaxin, G-CSF, GM-CSF, RANTES and MCP-1. Data are expressed as picograms per mL of BALF, mean ± SEM. Groups were compared to HDM/vehicle group using a one-way ANOVA, followed by a Dunnett's test;*P<0.05,*P<0.01,***P<0.001; n=8.*

BALF (pg/ml)	Saline/Saline /vehicle	HDM/Saline /vehicle	HDM/Poly I:C/vehicle	HDM/Poly I:C/1104 80µg/Kg i.v.	HDM/Poly I:C/Dex 1mg/kg p.o.
IFN-ɣ	25±10***	377±39	611±41##	396±33***	570±33
IL-4	20±8***	511±37	578±32	444±30*	498±31
IL-5	19±7***	407±31	621±28###	361±28***	478±29**
IL-6	18±7***	395±32	692±39###	461±39***	555±44*
IL-10	10±6*	86±17	74±20	74±16	77±19
KC	20±8***	318±28	594±30###	425±38***	562±30
IL-12p40	19±8***	259±25	274±30	201±25	265±24
IL-13	14±7***	337±25	626±31###	303±31***	563±33
IL-17	20±8***	85±8	102±17	90±10	95±13
Eotaxin	15±7***	288±30	325±36	177±24**	310±29
G-CSF	15±7***	210±21	329±23##	245±23	285±27
GM-CSF	13±7***	202±24	222±20	186±19	231±25
RANTES	22±9***	37±12	98±12##	82±8	91±10
MCP-1	21±8***	244±23	563±32###	369±25***	564±30

Table 2– Effect of treatment on cytokines/chemokines in BALF after HDM challenge and Poly I:C exacerbation. *Effect on IFN-ɣ, IL-4, IL-5, IL-6, KC, IL-10, IL-12(p40), IL-13, IL-17, Eotaxin, G-CSF, GM-CSF, RANTES and MCP-1. Data are expressed as picograms per mL of BALF, mean ±SEM. Groups were compared to HDM/Poly I:C/vehicle using a one-way ANOVA, followed by a Dunnett's test;*P<0.05, **P<0.01,***P<0.001. A Student's t-test was used when comparing HDM/saline/vehicle vs HDM/Poly I:C/vehicle;##P<0.01, ###P<0.001; n=8.*

Results (III)

Effect of treatment on Airway Hyperresponsiveness (AHR)

HDM challenge caused a significant increase in total resistance (RI) and decrease in dynamic compliance (Cdyn) which were exacerbated with Poly I:C administration (Fig4). Both 1104 and dexamethasone had a significant effect on RI and Cdyn in the absence of exacerbation (Fig4A,B) but only 1104 remained efficacious after exacerbation (Fig4B,D).

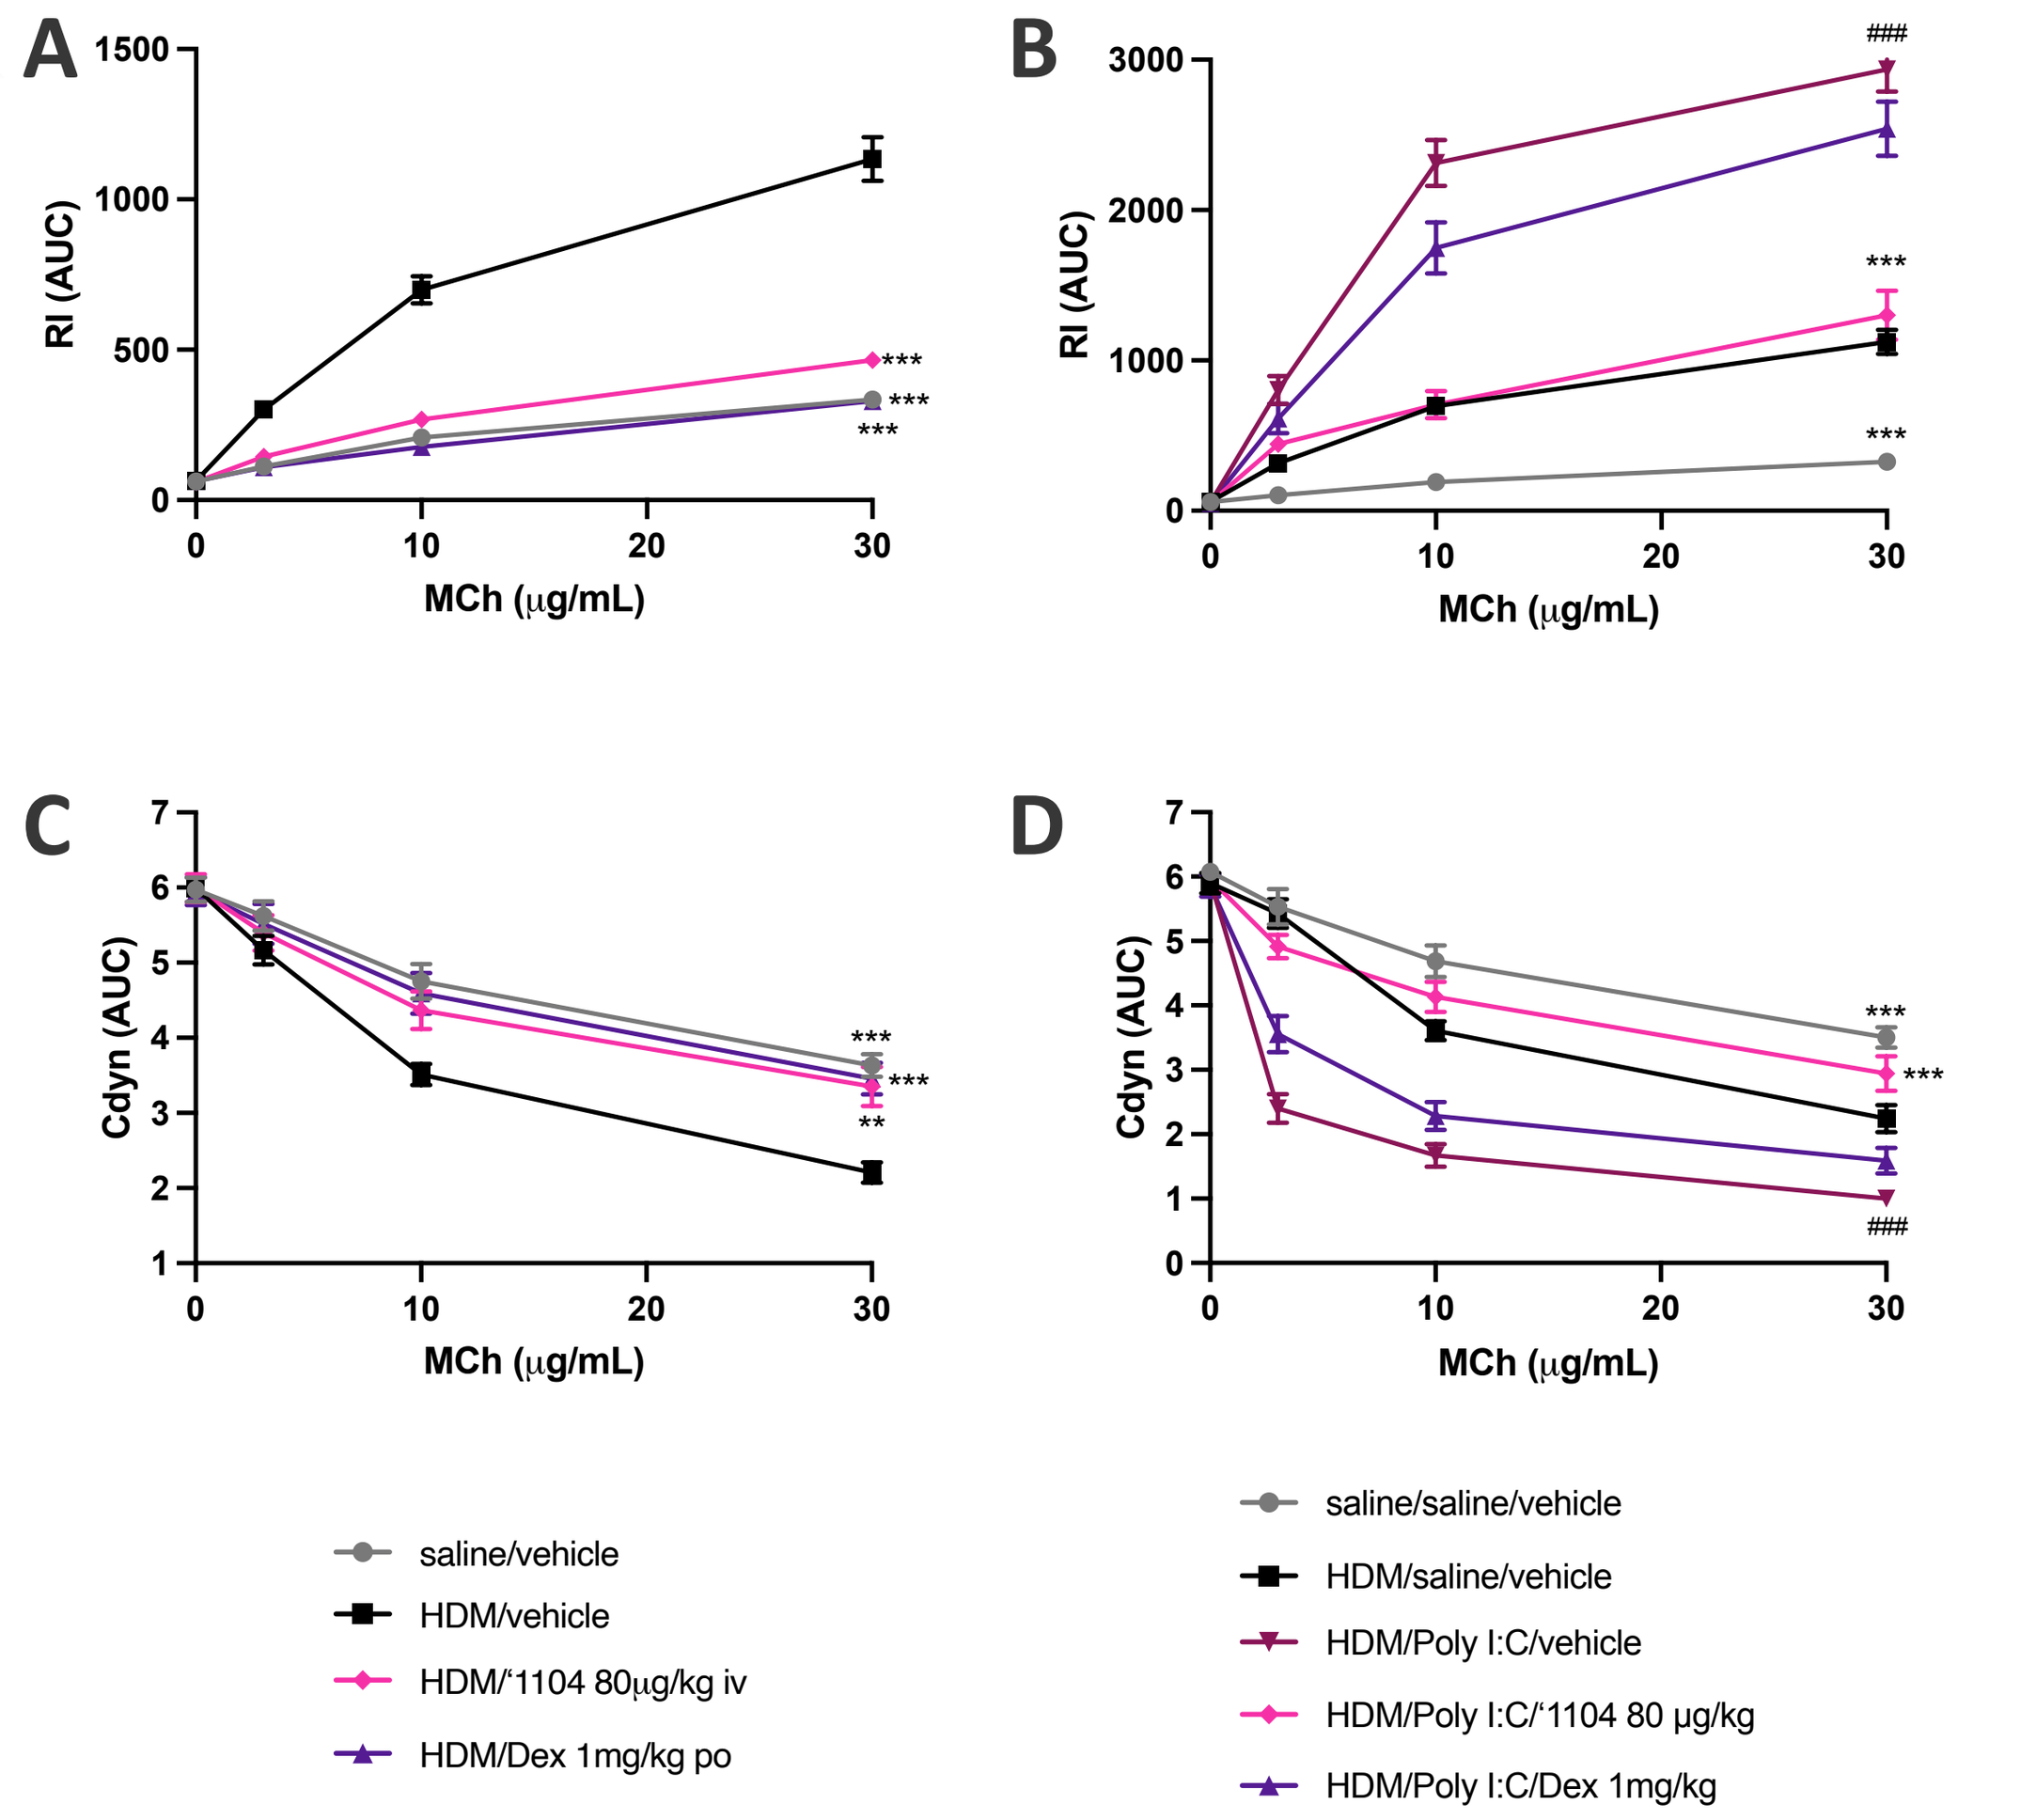
A
B
C
D
RI (AUC)
Cdyn (AUC)
MCh (µg/mL)
saline/vehicle
HDM/vehicle
HDM/'1104 80µg/kg iv
HDM/Dex 1mg/kg po
saline/saline/vehicle
HDM/saline/vehicle
HDM/Poly I:C/vehicle
HDM/Poly I:C/'1104 80 µg/kg
HDM/Poly I:C/Dex 1mg/kg

Figure 4. Effect of treatment on AHR to methacoline (MCh). *Effect of treatment on total lung resistance (RI) and dynamic compliance (Cdyn) in the absence (A,C) or presence (B,D) of Poly I:C exacerbation. Data are expressed as area under the curve (AUC), mean ±SEM. Groups were compared to either to HDM/vehicle (A,C) or HDM/Poly I:C/vehicle (B,D) groups using a one-way ANOVA, followed by a Dunnett's test;**P<0.01,***P<0.001. A Student's t-test was used when comparing HDM/saline/vehicle vs HDM/Poly I:C/vehicle group;###P<0.001; n=8.*

Conclusions

- IRL201104 is an immunomodulatory peptide that has shown efficacy in a recent Eosinophilic Esophagitis (EoE) phase 2A trial (NCT05084963; Dellon et al, 2023) The primary endpoint in the trial was met with multiple secondary positive findings. 1104 was recently granted orphan drug designation for EoE by the FDA and Revolo is planning a Phase IIb EoE study later this year.
- In this model of HDM driven allergic inflammation, with a mixed T cell background, 1104 showed a significant impact on inflammatory infiltration, HDM specific IgE, relevant cytokines/chemokines and lung function which was similar or better than positive control dexamethasone.
- As previously described, Poly I:C exacerbation rendered the model less sensitive to steroids. In the exacerbated model, unlike dexamethasone, 1104 largely maintained its efficacy on inflammatory infiltration, HDM specific IgE, relevant cytokines/chemokines and lung function.
- The present work shows the potential of IRL201104 in asthma and other allergic inflammatory diseases even in phenotypes that do not respond to steroids.

References

1. Page C et al. Am J Respir Crit Care Med 2019;199:A2861.
2. Riffo-Vasquez Y et al, Clin Exp Allergy. 2020 Apr;50(4):508-519.
3. De Alba J et al. Am J Respir Crit Care Med 2022; 205:A3743.
4. De Alba J et al. Clin Sci. 2015 Dec;129(11):973-87
5. Dellon et al, ACG 2023 Annual Scientific Meeting Abstracts. Vancouver, BC, Canada.

Acknowledgements

This study was carried out by Pharmidex Pharmaceutical Services Ltd (London) on behalf of Revolo Biotherapeutics Ltd.